Timing of initiation of neratinib after trastuzumab-based adjuvant therapy in early stage HER2+ hormone receptor-negative breast cancer: Exploratory analyses from the phase III ExteNET trial

Bent Ejlertsen,[1] Carlos Barrios,[2] Erhan Gokmen,[3] Nicholas Robert,[4] Noelia Martinez,[5] John Link,[6] Serafin Morales,[7] Manuel Ruiz Borrego,[8] Manish Bhandari,[9] Yoshiaki Rai,[10] Adam Luczak,[11] Takashi Fujita,[12] Alvin Wong,[13] Bin Yao,[14] Miguel Martin[15]

[1]Rigshospitalet, Copenhagen, Denmark; [2]Pontifical Catholic University of Rio Grande do Sul School of Medicine, Porto Alegre, Brazil; [3]Faculty of Medicine, Ege University, Izmir, Turkey; [4]Virginia Cancer Specialists, The US Oncology Network, Fairfax, VA, USA; [5]Hospital Universitario Ramón y Cajal, Madrid, Spain; [6]Breastlink Medical Group, Inc, Santa Ana, CA, USA; [7]Hospital Universitari Arnau de Vilanova de Lleida, Lleida, Spain; [8]Oncology Department, Hospital Universitario Virgen del Rocio, Seville, Spain; [9]Christ Hospital of Cincinnati, Cincinnati, OH, USA; [10]Hakuaikai Medical Corporation Sagara Hospital, Kagoshima City, Japan; [11]Aalborg Sygehus, Aalborg, Denmark; [12]Jichi Medical University Hospital, Tochigi, Japan; [13]Puma Biotechnology Inc., South San Francisco, CA, USA; [14]Puma Biotechnology Inc., Los Angeles, CA, USA; [15]Instituto de Investigación Sanitaria Gregorio Marañón, Centro de Investigación Biomédica en Red de Cáncer, GEICAM, Universidad Complutense, Madrid, Spain

#549

Introduction

- Neratinib (Puma Biotechnology Inc.), an irreversible tyrosine kinase inhibitor of HER1, HER2, and HER4,[1] is approved for the extended adjuvant treatment of patients with early-stage HER2-positive (HER2+) breast cancer after trastuzumab-based adjuvant therapy.[2]
- The pattern of recurrence in early-stage HER2+ breast cancer is influenced by hormone receptor (HR) status.
 - While the risk of relapse is relatively constant over time in patients with HR-positive (HR+) tumors, the risk of recurrence is initially higher in patients with HR-negative (HR–) disease and then approximates the risk in HR+ disease.[3]
 - Further, patients with HER2+ HR– breast cancer have a higher rate of aggressive features, including poor differentiation and extracapsular extension, compared to HER2+ HR+ disease.[4]
- Prespecified subgroup analyses of the ExteNET study showed greater benefit with neratinib in patients with HR+ versus HR– tumors, and in patients who initiated neratinib ≤12 months after completing trastuzumab.[2]
 - To better understand the temporal effects of neratinib on disease recurrences in patients with HR– disease, we performed exploratory analyses examining the impact on efficacy of the interval between prior trastuzumab to the start of neratinib in the HR– subpopulation.

Methods

Study design

- ExteNET is an ongoing, multicenter, randomized, double-blind, phase III trial (Clinicaltrials.gov identifier: NCT00878709).
- Randomization was stratified by HR status (HR+ *vs* HR–), prior trastuzumab (administered concurrently *vs* sequentially with chemotherapy), and nodal status (negative, 1–3 positive nodes, or ≥4 positive nodes).
- Approval of neratinib was based on the primary 2-year efficacy analysis which showed that neratinib significantly improved 2-year invasive disease-free survival (iDFS) in patients with early-stage HER2+ breast cancer (hazard ratio 0.67; 95% confidence interval [CI] 0.50–0.91; p=0.009),[2] 5-year efficacy analysis,[5] and safety analysis,[2] all of which have been reported previously.
- Patient follow-up is ongoing, with an analysis of overall survival planned after 248 events.

Patients

- Women (N=2840) aged 18 years or older with histologically confirmed stage 1–3c disease (amended to 2–3c disease) HER2+ breast cancer.
- Patients had completed 1 year of trastuzumab-based (neo)adjuvant therapy up to 2 years (amended to 1 year) previously.

HR status

- HR status was determined locally before trial entry and tumors were classified according to local criteria.
- HR+ disease was defined as estrogen receptor-positive and/or progesterone receptor positive; HR– disease was defined as estrogen receptor-negative and progesterone receptor-negative.

Treatment

- Patients were randomized to either oral neratinib 240 mg once daily or placebo for 1 year.
- Neratinib dose reductions (to 200, 160, and 120 mg/day) were permitted for toxicity.

Outcomes

- Endpoints considered in the present analysis:
 - iDFS, the primary endpoint, defined as time from randomization to first occurrence of invasive ipsilateral tumor recurrence, invasive contralateral breast cancer, local/regional invasive recurrence, distant recurrence or death from any cause.
 - Disease-free survival including ductal carcinoma in situ (DFS-DCIS), a secondary endpoint, defined as time from randomization to the first occurrence of a disease-free survival event or ductal carcinoma in situ.

Data analysis

- Data on disease recurrences were collected prospectively during years 1 and 2 following randomization, and retrospectively from medical records for 5 years after randomization by the treating institution, by protocol amendment.
- Efficacy outcomes were analyzed in the HR– subgroup, and also among HR– patients categorized according to the interval between completing trastuzumab and randomization (i.e. 0–6, and >6 months), where date of randomization was an approximation for the start date of neratinib therapy.
- Time-to-event endpoints were analyzed using a Cox proportional hazards model, and Kaplan Meier methods were used to estimate 5-year event-free survival rates.
- Data cutoff was March 1, 2017.

Results

Patients

- The ExteNET intent–to-treat (ITT) population comprised 2840 patients, 1209 (43%) of whom had HR– disease (neratinib, n=604; placebo, n=605).
- The HR– population closely reflected the ITT population and no notable differences were observed for any of the key baseline demographics and disease characteristics (Table 1).

Table 1. Baseline patient demographics and characteristics (HR– subgroup and ITT population)

	HR– subgroup		ITT population	
Characteristic	Neratinib (n=604)	Placebo (n=605)	Neratinib (n=1420)	Placebo (n=1420)
Median (range) age, years	54 (27–81)	54 (28–82)	52 (25–83)	52 (23–82)
Race, n (%)				
White	475 (79)	460 (76)	1165 (82)	1135 (80)
Asian	99 (16)	99 (16)	188 (13)	197 (14)
Black or African American	12 (2)	22 (4)	27 (2)	47 (3)
Other	18 (3)	24 (4)	40 (3)	41 (3)
Region, n (%)				
North America	216 (36)	202 (33)	519 (37)	477 (34)
Western Europe, Australia, and South Africa	200 (33)	213 (35)	487 (34)	532 (38)
Asia Pacific, Eastern Europe, and South America	188 (31)	190 (31)	414 (29)	411 (29)
Nodal status, n (%)				
Negative	148 (25)	148 (24)	335 (24)	336 (24)
1–3 positive nodes	271 (45)	270 (45)	664 (47)	664 (47)
≥4 positive nodes	185 (31)	187 (31)	421 (30)	420 (30)
Prior trastuzumab regimen, n (%)				
Concurrent	378 (63)	378 (62)	884 (62)	886 (62)
Sequential	226 (37)	227 (38)	536 (38)	534 (38)
Median (range) time from last trastuzumab to randomization, months	4.4 (0.4–26.9)	4.4 (0.3–24.3)	4.4 (0.2–30.9)	4.7 (0.3–40.6)
Prior neoadjuvant therapy, n (%)	152 (25)	162 (27)	342 (24)	379 (27)

Treatment exposure

- In patients with HR– disease, the median duration of treatment was 11.7 months (range 0.1–12.7) in the neratinib group and 11.8 months (range 0.2–13.0) in the placebo group.
- Adherence with treatment in patients with HR– disease, defined as the actual dose intensity as a percentage of the planned dose intensity, was 98% (standard deviation [SD] 7%) in the neratinib group and 99% (SD 3%) in the placebo group.

Efficacy outcomes

- The median follow-up duration in the HR– subgroup was 5.2 years.
- Five-year iDFS data in patients with HR– tumors, stratified by the interval between prior trastuzumab and randomization, are shown in Table 2 and corresponding Kaplan-Meier plots are shown in Figure 1.

Table 2. Analysis of iDFS at 5 years in patients with HR– tumors and by the interval between completion of prior trastuzumab and randomization

		5-year iDFS rate, %		
Interval between prior trastuzumab and randomization, months	n	Neratinib	Placebo	Hazard ratio (95% CI)[a]
0–6	695	88.9	86.1	0.73 (0.47–1.14)
>6	514	88.7	92.7	1.52 (0.82–2.88)
All patients with HR– disease		88.8	88.9	0.95 (0.66–1.35)

[a]Neratinib *vs* placebo.

Figure 1. Kaplan–Meier plots of iDFS at 5 years in patients with HR– tumors and by the interval between completion of prior trastuzumab and randomization

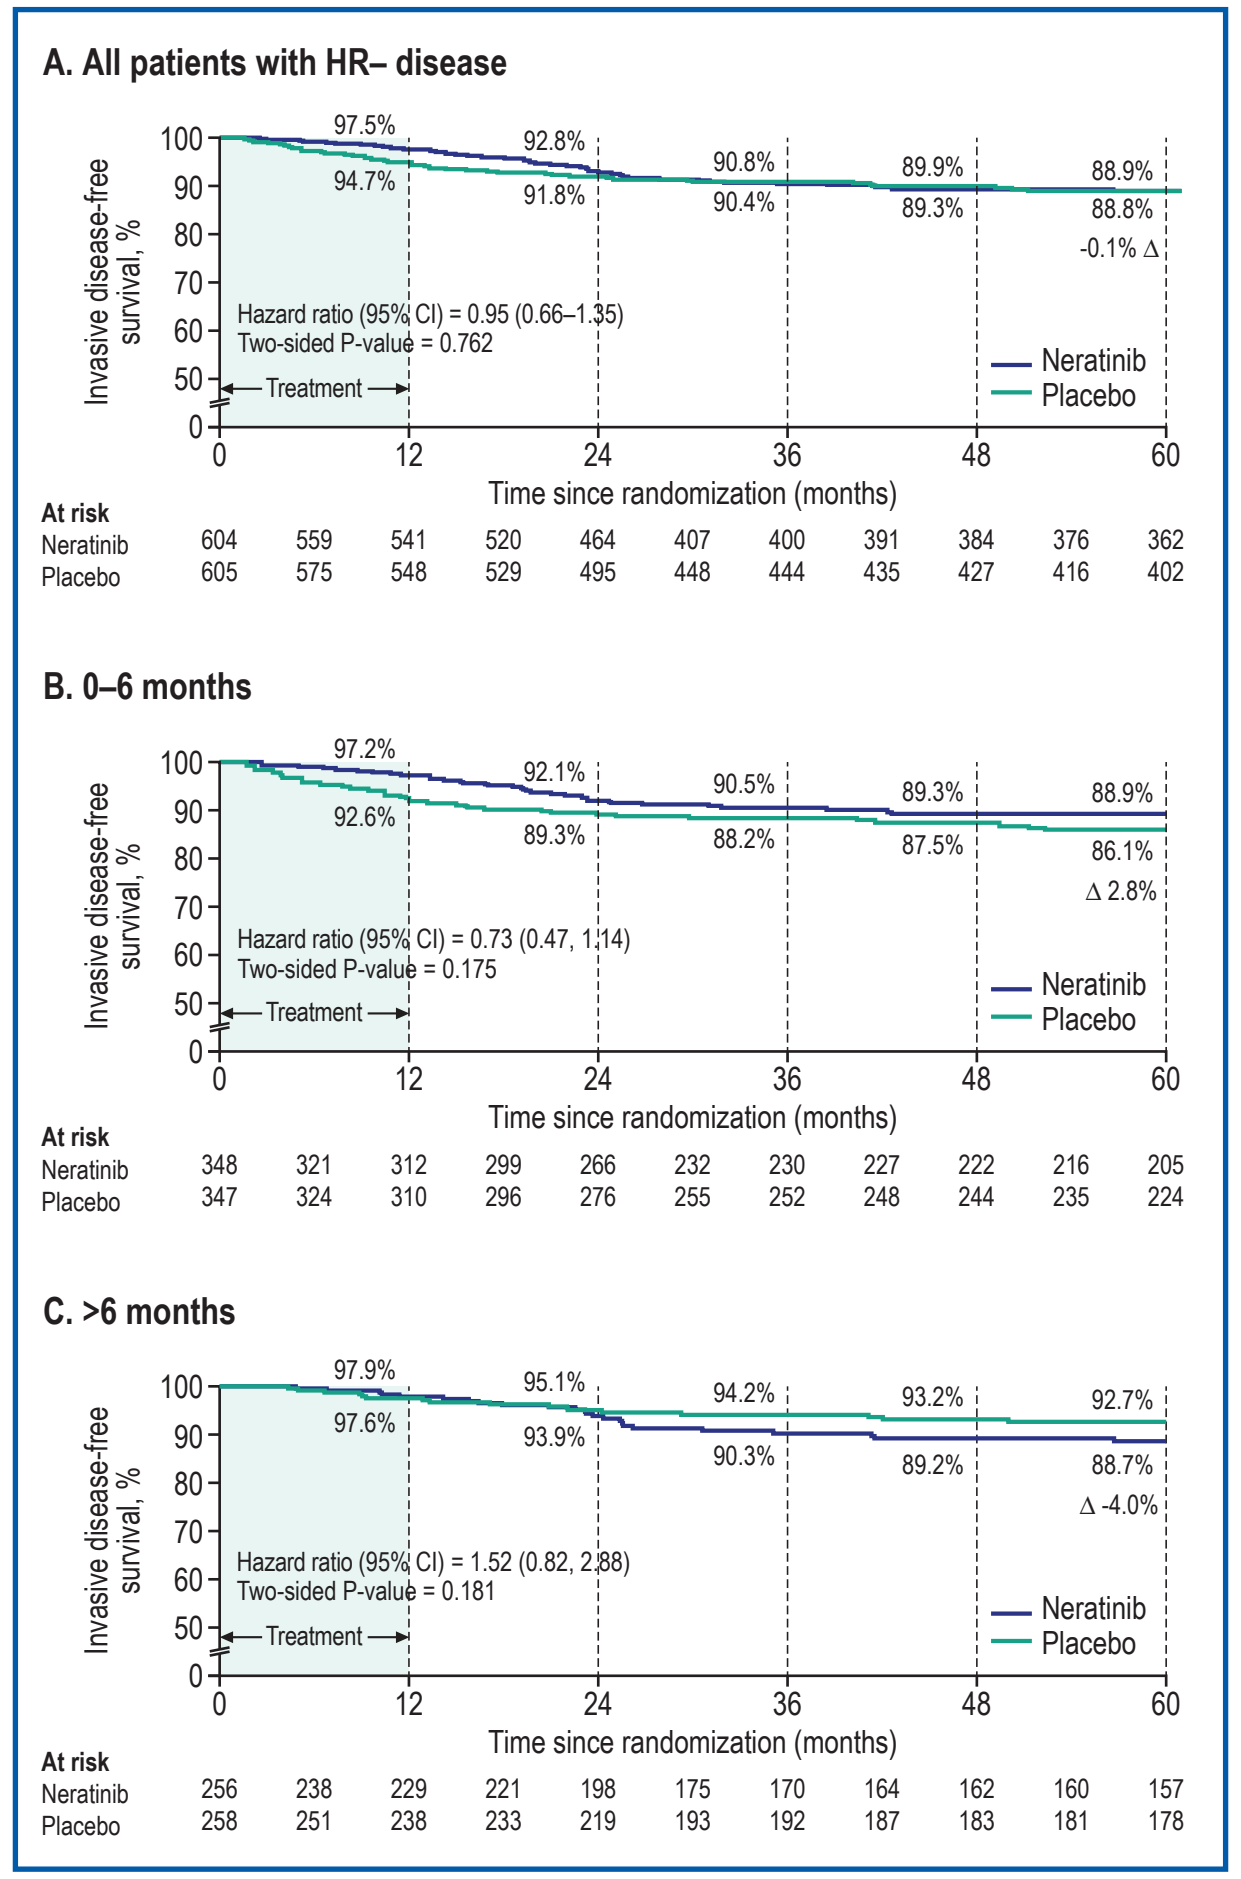
A. All patients with HR– disease
Invasive disease-free survival, %
97.5%
94.7%
92.8%
91.8%
90.8%
90.4%
89.9%
89.3%
88.9%
88.8%
-0.1% Δ
Hazard ratio (95% CI) = 0.95 (0.66–1.35)
Two-sided P-value = 0.762
Treatment
Neratinib
Placebo
Time since randomization (months)
At risk
Neratinib 604 559 541 520 464 407 400 391 384 376 362
Placebo 605 575 548 529 495 448 444 435 427 416 402
B. 0–6 months
97.2%
92.6%
92.1%
89.3%
90.5%
88.2%
89.3%
87.5%
88.9%
86.1%
Δ 2.8%
Hazard ratio (95% CI) = 0.73 (0.47, 1.14)
Two-sided P-value = 0.175
At risk
Neratinib 348 321 312 299 266 232 230 227 222 216 205
Placebo 347 324 310 296 276 255 252 248 244 235 224
C. >6 months
97.9%
97.6%
95.1%
93.9%
94.2%
90.3%
93.2%
89.2%
92.7%
88.7%
Δ -4.0%
Hazard ratio (95% CI) = 1.52 (0.82, 2.88)
Two-sided P-value = 0.181
At risk
Neratinib 256 238 229 221 198 175 170 164 162 160 157
Placebo 258 251 238 233 219 193 192 187 183 181 178

All HR– patients

- At the 5-year cutoff, the iDFS rate in patients with HR– disease was similar in both groups: 89% in the neratinib group and 89% in the placebo group (hazard ratio 0.95; 95% CI 0.66–1.35; two-sided p=0.762).
 - Neratinib elicited a transient beneficial effect on iDFS in patients with HR– disease that diminished after treatment was completed (Figure 1A).
 - Observations from the placebo group suggested that the risk of recurrence was greater in patients with HR– tumors who had recently completed trastuzumab-based therapy (Figure 1A). The risk of recurrence then diminished considerably at later time-points after completion of trastuzumab.
- Similar trends were observed with respect to DFS-DCIS, a secondary efficacy endpoint (Table 3).

Table 3. Analysis of DFS-DCIS at 5 years in patients with HR– tumors and by the interval between completion of prior trastuzumab and randomization

		5-year DFS-DCIS rate, %		
Interval between prior trastuzumab and randomization, months	n	Neratinib	Placebo	Hazard ratio (95% CI)[a]
0–6	695	88.1	85.4	0.75 (0.48–1.15)
>6	514	87.6	91.4	1.40 (0.78–2.53)
All patients with HR– disease	1209	87.9	87.9	0.94 (0.66–1.32)

[a]Neratinib *vs* placebo.

HR– patients by interval between prior trastuzumab and randomization

- When patients with HR–disease were analyzed by the interval between prior trastuzumab and randomization, the hazard ratio in the neratinib group compared with the placebo group was 0.73 (0.47–1.14) when the interval was 0–6 months, and 1.52 (0.82–2.88) when >6 months (Table 2, Figure 1B and 1C).
 - Similar trends were observed with respect to DFS-DCIS (Table 3).

Conclusions

- The risk of recurrence in the population with HR– HER2+ early-stage breast cancer was greatest during the first 6 months following completion of trastuzumab and then decreased over time.
- The greatest benefit with neratinib among HR– patients was noted in the subgroup with the highest risk of relapse, i.e. those completing trastuzumab within 6 months of randomization.
- The findings from these exploratory analyses are hypothesis-generating only and need to be replicated in further studies.

References

1. Rabindran SK. Cancer Lett 2005;227:9–23.
2. Chan A, et al. Lancet Oncol 2016;17:367–77.
3. Untch M, et al. Ann Oncol 2008;19:1090–6.
4. Gomez HL, et al. Hematol Oncol Stem Cell Ther 2010;3:109–15.
5. Martin M, et al. Lancet Oncol 2017;18:1688–1700.

Acknowledgements

ExteNET was sponsored by Wyeth, Pfizer, and Puma Biotechnology Inc. Puma Biotechnology Inc. also funded the provision of editorial support provided by Miller Medical Communications.

Copyright 2018 Puma Biotechnology

Presented at the American Society of Clinical Oncology (ASCO) Annual Meeting, June 1–5, 2018, in Chicago, IL, USA